An Improved Performance Adsorbent for the Downstream Processing of Serum Albumin and Related Fusion Proteins

Bastiaan Lobbezoo, Ben Galarza, Jim Morritt, Chloe Booth, Jennifer Walden and Dev Baines

ProMetic BioSciences Ltd., 211 Cambridge Science Park, Cambridge, CB4 0WA UK

Introduction

Affinity chromatography is ideally suited to the purification of biopharmaceuticals due to the unique specificity of affinity ligands for the target protein. Thus, highly selective separation based on affinity chromatography in packed bed columns incorporating capture, concentration and a high degree of purification provides a simple and effective means of reducing the number of processing steps thereby removing some of the perceived bottlenecks in downstream processing.

Significant improvements in reducing the manufacturing costs are possible and can be achieved by shortening processing times by increasing flow rates through chromatography matrix optimisation with concomitant ability to clean and re-use the chromatography adsorbent.

Albumin-fusion protein technology represents an increasingly important alternative platform for the production of therapeutically significant proteins with extended *in vivo* half-lives. A number of proteins and bioactive peptides fused to albumin are currently being investigated for use in therapeutic applications.

Mimetic Blue® SA ligand is highly selective for albumin and affinity chromatography adsorbents based on this ligand provide a platform technology for the purification of human albumin and genetically engineered albumin-fusion proteins. The existing adsorbents are based on a standard 6% cross-linked agarose. Optimisation of the cross-linking chemistry of the PuraBead® 6 base matrix (a 6% near-monodisperse beaded agarose), with optimal coupling of Mimetic Blue SA ligand has led to the development of a new adsorbent with improved performance (described below).

The resulting adsorbent Mimetic Blue SA HL P6HF retains the high binding capacity (~30g albumin/L) seen for the existing Mimetic Blue SA products and is characterised by significant (3 fold) improvement in the pressure flow properties. One-step purifications of human serum albumin from plasma and an albumin-fusion protein are described.

Pressure versus flow

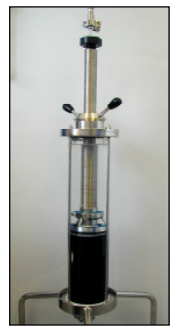

- Column – BPG 100
- Column diameter – 10 cm
- Bed height – 20 cm
- Column volume (CV) – 1 L
- Buffer – 50 mM sodium phosphate, 0.1 M NaCl, pH 7.5

Figure 1: Column chromatography apparatus to perform pressure versus flow for Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

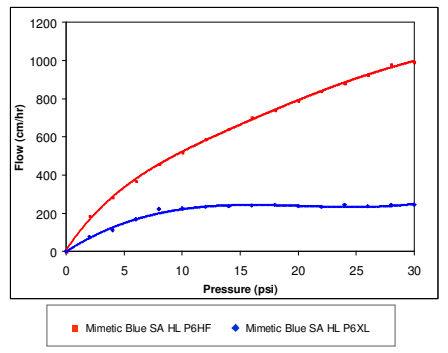

Flow (cm/hr)
Pressure (psi)
Mimetic Blue SA HL P6HF
Mimetic Blue SA HL P6XL

Figure 2: Pressure versus flow results for Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

Binding capacity versus flow

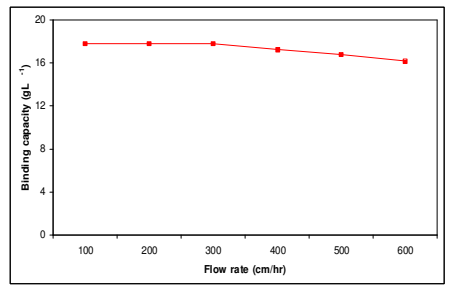

Binding capacity (gL⁻¹)
Flow rate (cm/hr)

Figure 3: Human serum albumin binding capacity results for Mimetic Blue SA HL P6HF loading **human source plasma** to breakthrough from 100 to 600 cm/hr (6 to 1 minute residence time), using a 2.6 cm diameter column with a 10 cm bed height (53 mL CV).

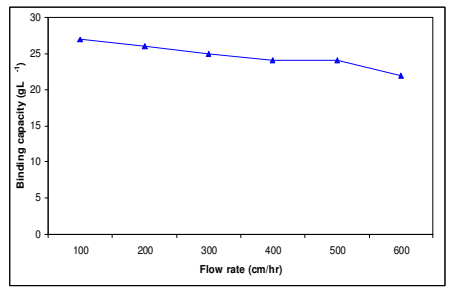

Binding capacity (gL⁻¹)
Flow rate (cm/hr)

Figure 4: Human serum albumin binding capacity results for Mimetic Blue SA HL P6HF loading **purified HSA** to breakthrough from 100 to 600 cm/hr (6 to 1 minute residence time), using a 2.6 cm diameter column with a 10 cm bed height (53 mL CV).

Human serum albumin purification

Platform	Automated Chromatography Workstation
Column Conditions	11.6 mL column volume (1 cm diameter, 14.8 cm bed height)
Packing Flow Rate	600 cm/hr *
Operational Flow Rates	300 cm/hr **
Loading Flow Rate	150 cm/hr (6 min residence time)
Equilibration Buffer	50 mM sodium phosphate, pH 6.0
Load	7 mL human source plasma
Elution Buffer	50 mM sodium phosphate, 100 mM NaCl, 30 mM caprylate, pH 6.0
CIP (Clean in Place)	0.5 M NaOH

* Packing flow rate for Mimetic Blue SA HL P6XL – 450 cm/hr
** Operational flow rates for Mimetic Blue SA HL P6XL – 200 cm/hr

Table 1: Chromatography conditions for the capture and recovery of human serum albumin from human source plasma using Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

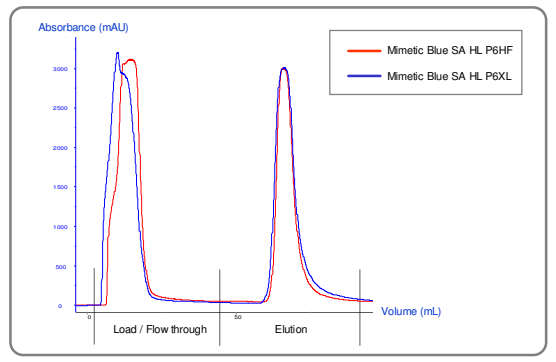

Absorbance (mAU)
Mimetic Blue SA HL P6HF
Mimetic Blue SA HL P6XL
Volume (mL)
Load / Flow through
Elution

Figure 5: Chromatogram overlays for the capture and recovery of human serum albumin from human source plasma using Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

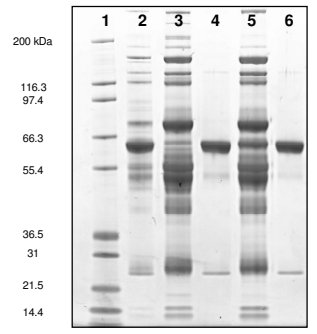

1 2 3 4 5 6
200 kDa
116.3
97.4
66.3
55.4
36.5
31
21.5
14.4

Lane 1 – Mw marker
Lane 2 – Load
Lane 3 – FT (**1**)
Lane 4 – Elution (**1**)
Lane 5 – FT (**2**)
Lane 6 – Elution (**2**)

(**1**) – Mimetic Blue SA HL P6XL
(**2**) – Mimetic Blue SA HL P6HF

Figure 6: Reduced SDS–PAGE analysis of flow through (FT) and elution chromatography fractions for Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL from human source plasma.

Adsorbent	Dynamic Binding Capacity (gL⁻¹)	Recovery (gL⁻¹)
Mimetic Blue SA HL P6XL	**>16**	**17.8**
Mimetic Blue SA HL P6HF	**>16**	**16.9**

Table 2: Human serum albumin binding capacity and recovery (Nephelometry) results from human source plasma for Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

Albumin-fusion protein purification

Platform	Automated Chromatography Workstation
Column Conditions	4.0 mL column volume (1 cm diameter, 5.0 cm bed height)
Packing Flow Rate	450 cm/hr
Operational Flow Rates	200 cm/hr
Loading Flow Rate	75 cm/hr (4 min residence time)
Equilibration Buffer	50 mM sodium phosphate, 25 mM NaCl, pH 7.0
Load	10 mL albumin-fusion protein feedstock
Elution Buffer	50 mM sodium phosphate, 25 mM NaCl, 30 mM caprylate, pH 7.0
CIP (Clean in Place)	0.5 M NaOH

Table 3: Chromatography conditions for the capture and recovery of albumin-fusion protein using Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

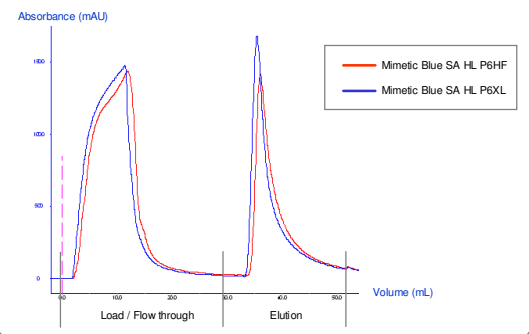

Absorbance (mAU)
Mimetic Blue SA HL P6HF
Mimetic Blue SA HL P6XL
Volume (mL)
Load / Flow through
Elution

Figure 7: Chromatogram overlays for the capture and recovery of albumin-fusion protein using Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

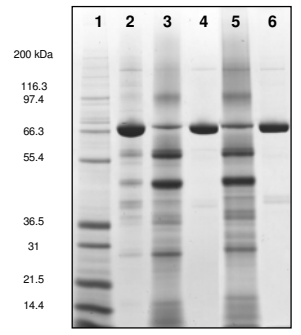

1 2 3 4 5 6
200 kDa
116.3
97.4
66.3
55.4
36.5
31
21.5
14.4

Lane 1 – Mw marker
Lane 2 – Load
Lane 3 – FT (**1**)
Lane 4 – Elution (**1**)
Lane 5 – FT (**2**)
Lane 6 – Elution (**2**)

(**1**) – Mimetic Blue SA HL P6HF
(**2**) – Mimetic Blue SA HL P6XL

Figure 8: Non-reduced SDS–PAGE analysis of flow through (FT) and elution chromatography fractions for albumin-fusion protein using Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

Adsorbent	Dynamic Binding Capacity (gL⁻¹)	Recovery (gL⁻¹)
Mimetic Blue SA HL P6XL	**>10.5**	**9.75**
Mimetic Blue SA HL P6HF	**>10.5**	**9.15**

Table 4: Albumin-fusion protein binding capacity and recovery (Nephelometry) results for Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL.

Adsorbent development

Adsorbent	Dynamic Binding Capacity (gL⁻¹)	
Mimetic Blue SA HL P6XL	**33**	
Mimetic Blue Development Batch (1)	**19**	Low Ligand Density
Mimetic Blue Development Batch (2)	**29**	
Mimetic Blue Development Batch (3)	**34**	
Mimetic Blue Development Batch (4)	**32**	
Mimetic Blue Development Batch (5)	**37**	High Ligand Density

Table 5: Binding data (A280) for Mimetic Blue SA HL P6XL and five Mimetic Blue SA HL P6HF development batches, using **purified HSA** (11.8 mL CV, 1 cm diameter, 15 cm bed height).

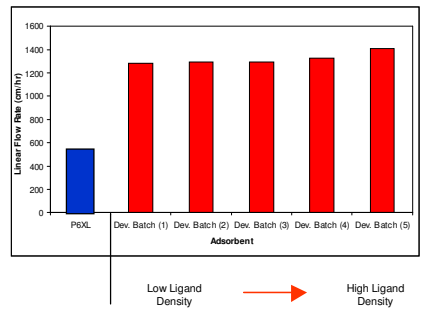

Linear Flow Rate (cm/hr)
P6XL
Dev. Batch (1)
Dev. Batch (2)
Dev. Batch (3)
Dev. Batch (4)
Dev. Batch (5)
Adsorbent
Low Ligand Density
High Ligand Density

Figure 9: Maximum linear flow rate results (150 mL CV, 15 cm bed height and 3.5 cm diameter) for Mimetic Blue SA HL P6XL and five Mimetic Blue SA HL P6HF development batches.

Adsorbent	Dynamic Binding Capacity (gL⁻¹)	Recovery (gL⁻¹)
Mimetic Blue SA HL P6XL	**>10.5**	**9.75**
Mimetic Blue Development Batch (2)	**>10.5**	**9.15**
Mimetic Blue Development Batch (3)	**>10.5**	**7.25**

Table 6: Binding capacity and recovery (Nephelometry) results for Mimetic Blue SA HL P6XL and two Mimetic Blue SA HL P6HF development batches, using **albumin-fusion protein feedstock** (4 mL CV, 1 cm diameter, 5 cm bed height).

Adsorbent	Dynamic Binding Capacity (gL⁻¹)	Recovery (gL⁻¹)
Mimetic Blue SAHL P6HF Verification Batch (1)	**>21.5**	**21.2**
Mimetic Blue SAHL P6HF Verification Batch (2)	**>20.3**	**20.5**
Mimetic Blue SAHL P6HF Verification Batch (3)	**>18.0**	**18.2**

Table 7: Binding capacity and recovery (Nephelometry) results for three Mimetic Blue SA HL P6HF verification batches, using **human source plasma** (11.8 mL CV, 1 cm diameter, 15 cm bed height).

Conclusions

- Mimetic Blue SA HL P6HF has a column flow rate of 1000 cm/hr in comparison to ~200 cm/hr for Mimetic Blue SA HL P6XL at 30 psi using a 10 cm diameter column.
- Mimetic Blue SA HL P6HF shows no significant decrease in human albumin dynamic binding capacity loading from 100 to 600 cm/hr (6 to 1 minute residence time), using both purified HSA and human source plasma.
- Both Mimetic Blue SA HL P6HF and Mimetic Blue SA HL P6XL show comparable dynamic binding capacity, recovery and purity results using albumin-fusion protein and human source plasma feedstocks.
- The final Mimetic Blue SA HL P6HF adsorbent was selected based on comparable purification and improved chromatography performance to Mimetic Blue SA HL P6XL.

Acknowledgements

The help of Steve Burton, Stuart Jordan, Sharon Williams and Ben Beacom at ProMetic has been greatly appreciated in the successful completion of this work.

IBC's BioProcess International™ Conference & Exhibition; October 12-16, 2009

Mimetic Blue, PuraBead and the PBL logo are registered trademarks of ProMetic BioSciences Ltd. © ProMetic BioSciences Ltd. 2009